Investigation of an urban farm intervention for a low income Hispanic population with multiple risk factors for diabetes

Monique Centrone Stefani,[1] Carrie Durward,[2] Rebecca Kline,[3] Debbie Humphries[4]

[1]Department of Sociology and Social Research, Università di Trento, Trento, Italy [2]Department of Nutrition, Dietetics, and Food Science, Utah State University, Logan, UT, [3]New Haven Farms, New Haven, CT, [4]Yale School of Public Health, New Haven, CT

Abstract

Program Number: 624.3

Obesity and chronic disease are highly prevalent in the United States, particularly among low-income Hispanic populations. Innovative approaches to changing social, behavioral and nutritional risk factors are crucially needed. This research evaluates the New Haven Farms Fresh Produce Prescription program, which provides regular exposure to and participation in urban agriculture to impact the consumption of fruits and vegetables as well as diabetes risk factors. Program members, an economically disadvantaged Hispanic population at risk for diabetes, were recruited from the Fair Haven Community Health Center, in New Haven, CT. Criteria for inclusion is based on the presence of at least two diabetes risk factors and income within 200% of the 2010 U.S. Poverty line. The intervention consists of a sixteen-week program that engages subjects in weekly visits to a farm in Fair Haven, CT where they receive a cooking and nutrition demonstration that focuses on fresh vegetables harvested that week from the farm and supplied to members. Subjects also learn skills for planting their own vegetables. Pre/Post-test includes standard anthropometric measurements, Spanish translations of a national validated dietary screener to estimate fruit and vegetable intake (NHANES Dietary Screener) and food security status (the USDA Household Food Security Survey Module), a risk-factor for obesity and diabetes. With this research, New Haven Farms' model is examined as a food system approach to impact obesity for those with multiple risk factors.

Introduction

Prevalence of obesity and diabetes in the U.S. are of epidemic proportions. With many theories regarding the causes of the obesity epidemic, there is increasingly the perspective that fewer people involved in, and aware of, how food is grown is a relevant factor. Several different studies have explored how engaging with agricultural production changes perspectives on food and food behaviors. However, studies of the effects of engaging in urban agriculture on food consumption and chronic disease risk factors are limited.

Context: New Haven is a post-industrial, urban community, with high levels of food insecurity, diabetes, and obesity.

- Seventh most food insecure of the 169 towns in Connecticut
- New Haven County has highest food insecurity in the state (13.6%).
- Obesity rate is 26.7%
- Diabetes prevalence is 8.3%.

Approach: New Haven Farms is a multi-level urban farm intervention to reduce nutrition related risk of diabetes in a low-income, primarily Hispanic community.

Hypotheses:

1: Fresh Produce Prescription Program membership improves food security status.
2: Fresh Produce Prescription Program membership improves fruit and vegetable intake.
3: Fresh Produce Prescription Program positively impacts diabetes risk factors.

Participation in community gardens and community supported agriculture may increase fruit and vegetable intake.

- 56% of community gardeners report eating 5-a-day vs. 25% of non-gardeners (Litt et al. 2011)
- Adults in gardening households were 3.5 times more likely to consume 5 fruits or vegetables per day (Alaimo et al. 2008)
- Low-income community garden members in Toronto reported that gardening improved nutrition through increased vegetable intake. (Wakefield et al. 2007)
- CSA members consume more dark green and yellow vegetables (Cohen et al, 1997)
- Market basket delivery increases vegetable intake by 1 serving per day in seniors (Johnson et al, 2004)

Nutrition education can improve diet and health outcomes in low-income population and reduce risk of diabetes.

- A combination of education and farmers market coupons is more effective in increasing intake than either alone (Anderson et al, 2001)

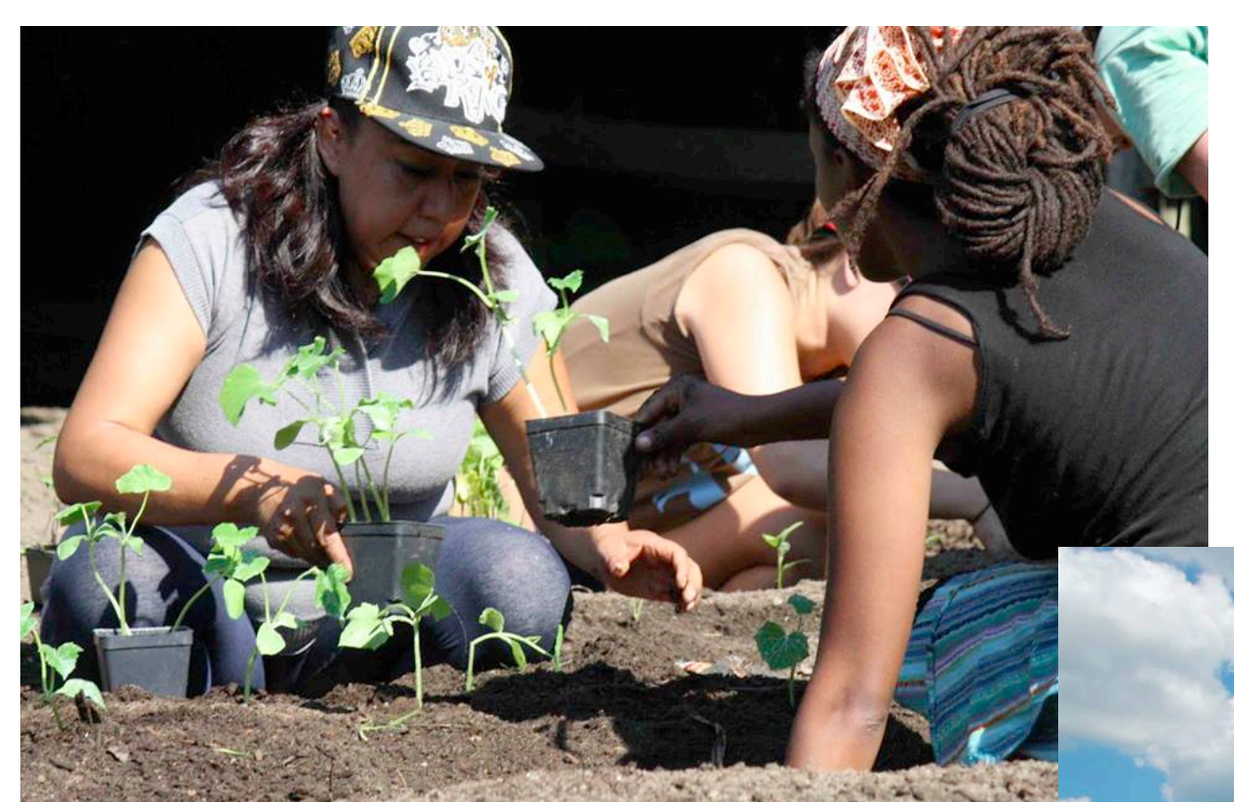
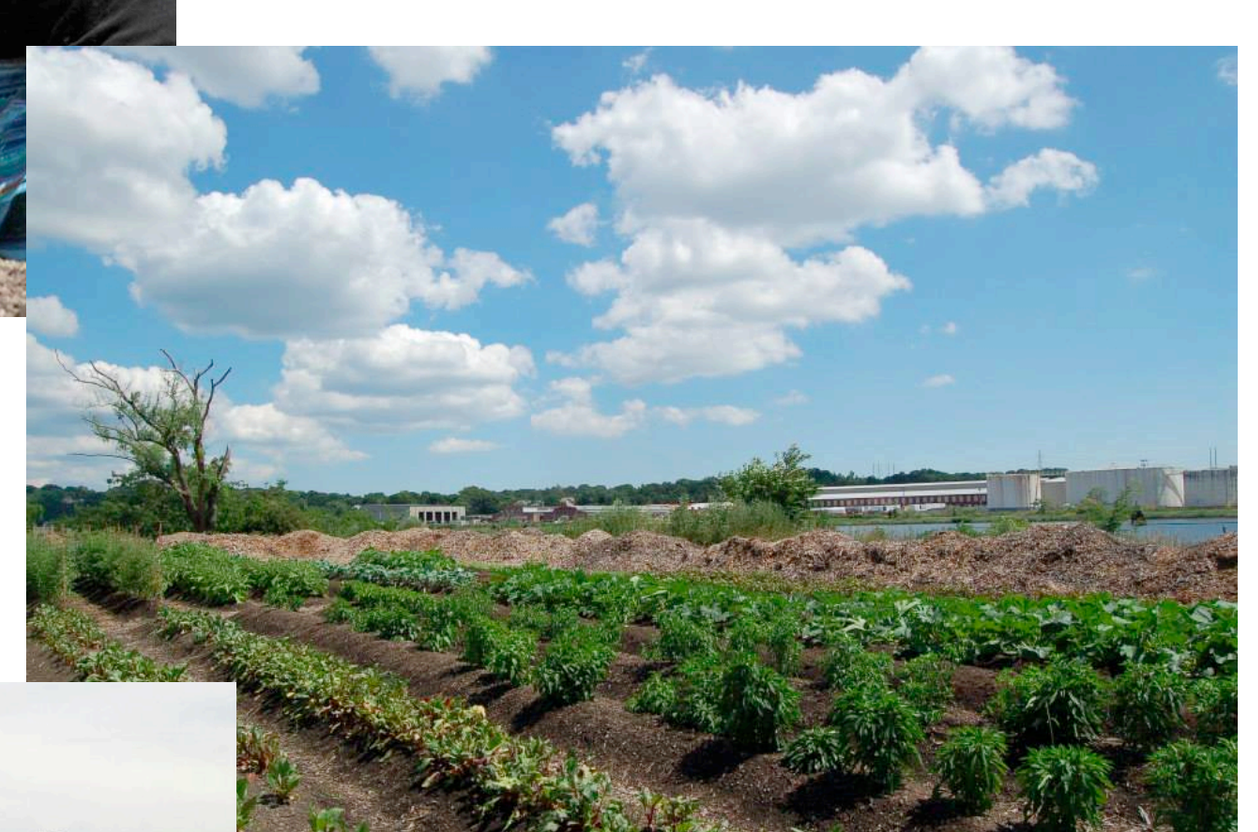
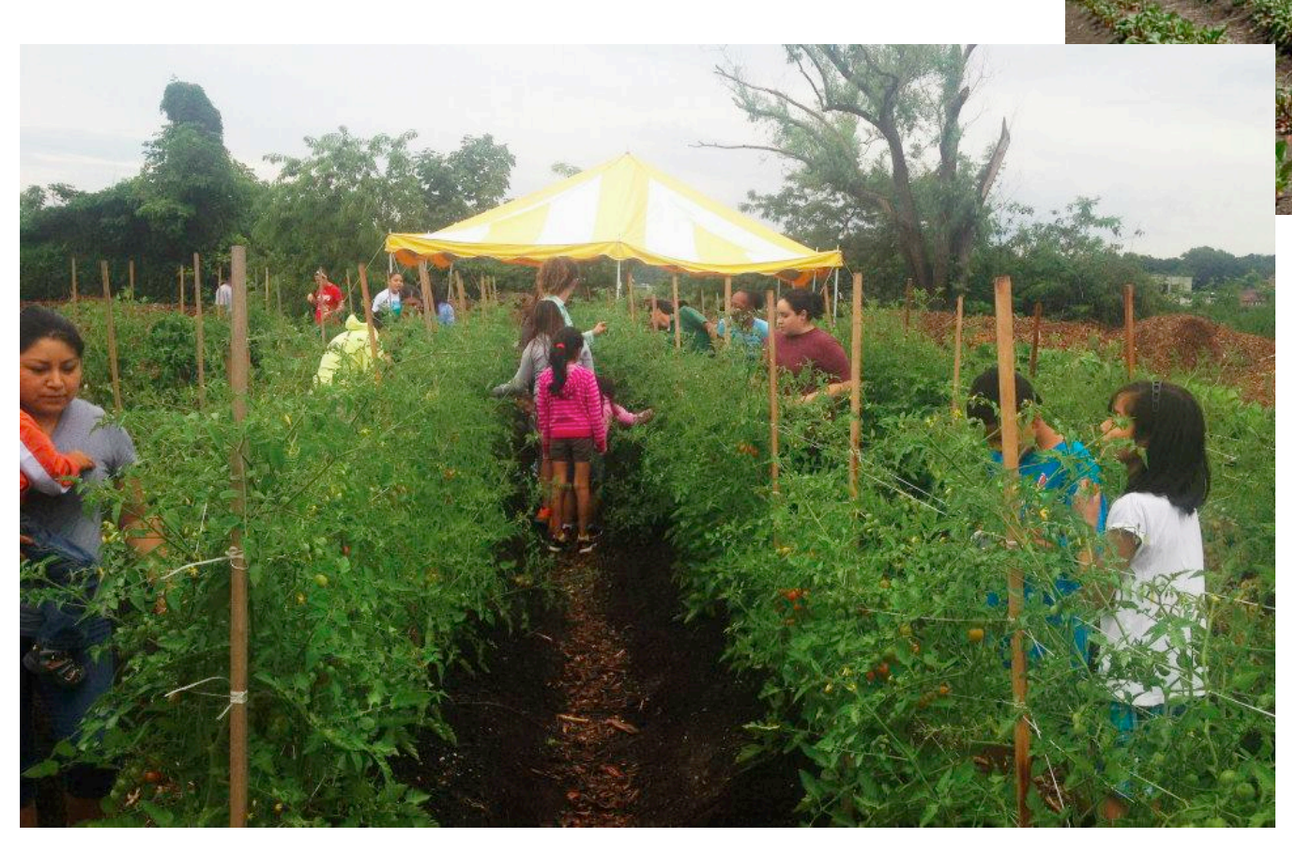

Contact Information
monique.centronestefani@gmail.com
carrie.durward@usu.edu
Debbie.humphries@yale.edu
executivedirector@newhavenfarms.org

Methods

Eligibility: (a) income within 200% of the federal poverty level and (b) two or more risk factors for diabetes.

Recruitment: Participants were recruited through referral from clinicians at a federally qualified community health center in New Haven, CT

Measurement: Food security status assessed pre/post intervention using USDA HFSSM; dietary intake used NHANES DSQ; Anthropometry pre/post intervention; Weekly survey to report on use of previous week's food basket.

Intervention/Treatment: The 16-week comprehensive urban farm intervention included weekly nutrition, cooking, and gardening education and a weekly share of produce.

Weekly Produce Shares: On average, the weekly shares contained 38.5 cups of raw, cleaned, ready to eat produce. Vegetables made up a majority of the produce (86%) with fruit making up 14% of the produce over the entire season. Per day, the shares provided 5.5 cups of vegetables for the duration of the intervention.

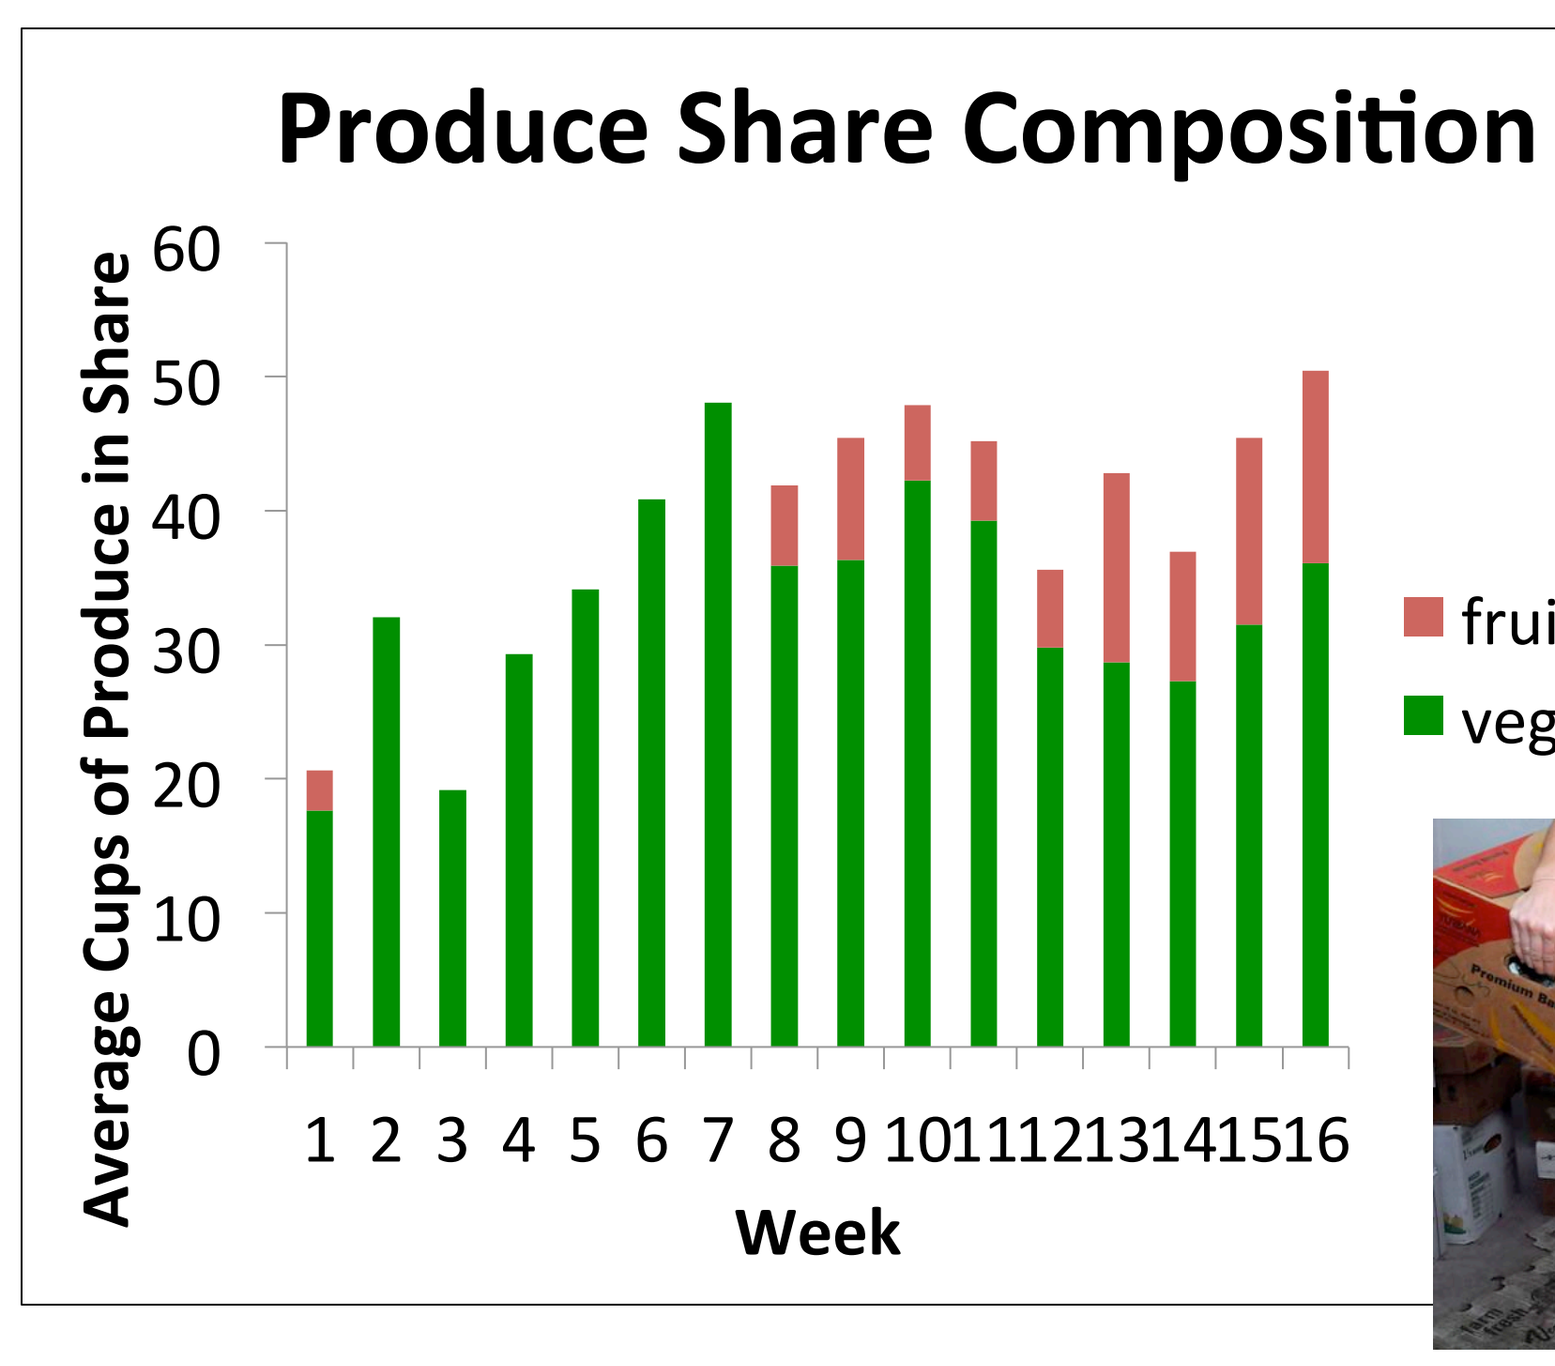
Produce Share Composition
Average Cups of Produce in Share
0
10
20
30
40
50
60
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16
Week
fruit
vegetable

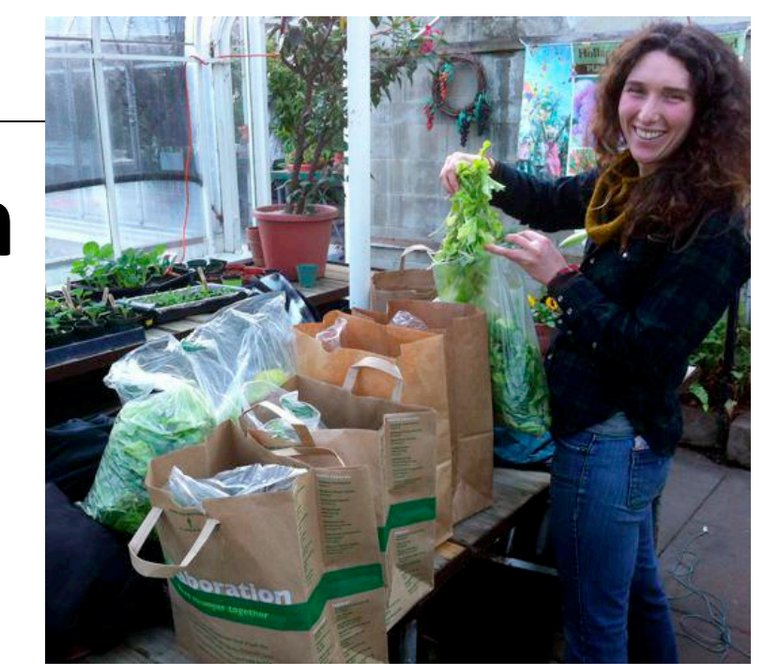
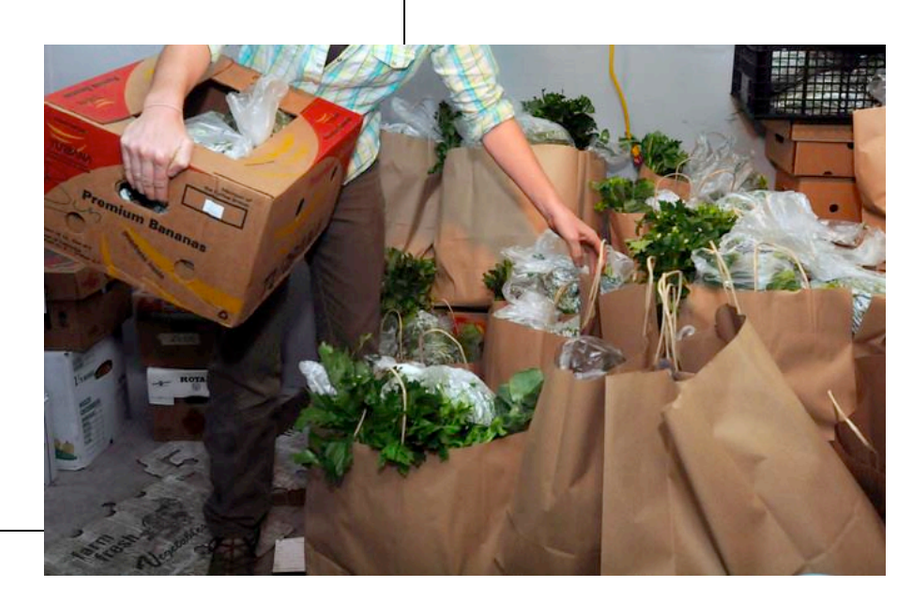

Weekly Education Sessions:

- Cooking and tasting demonstrations by a professional chef
- Nutrition education based on MyPlate and USDA 10-tips materials. Topics included food groups, portion sizes, eating better on a budget, food safety, and other fruit and vegetable promotion topics.
- Hands-on gardening education – weeding, planting, harvesting

On average, participants attended 11 education sessions.

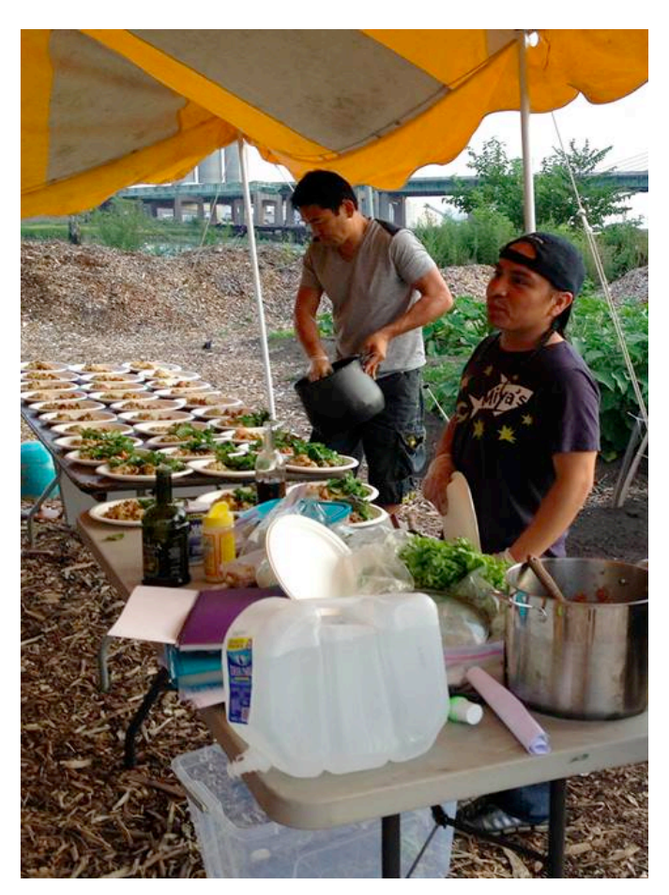
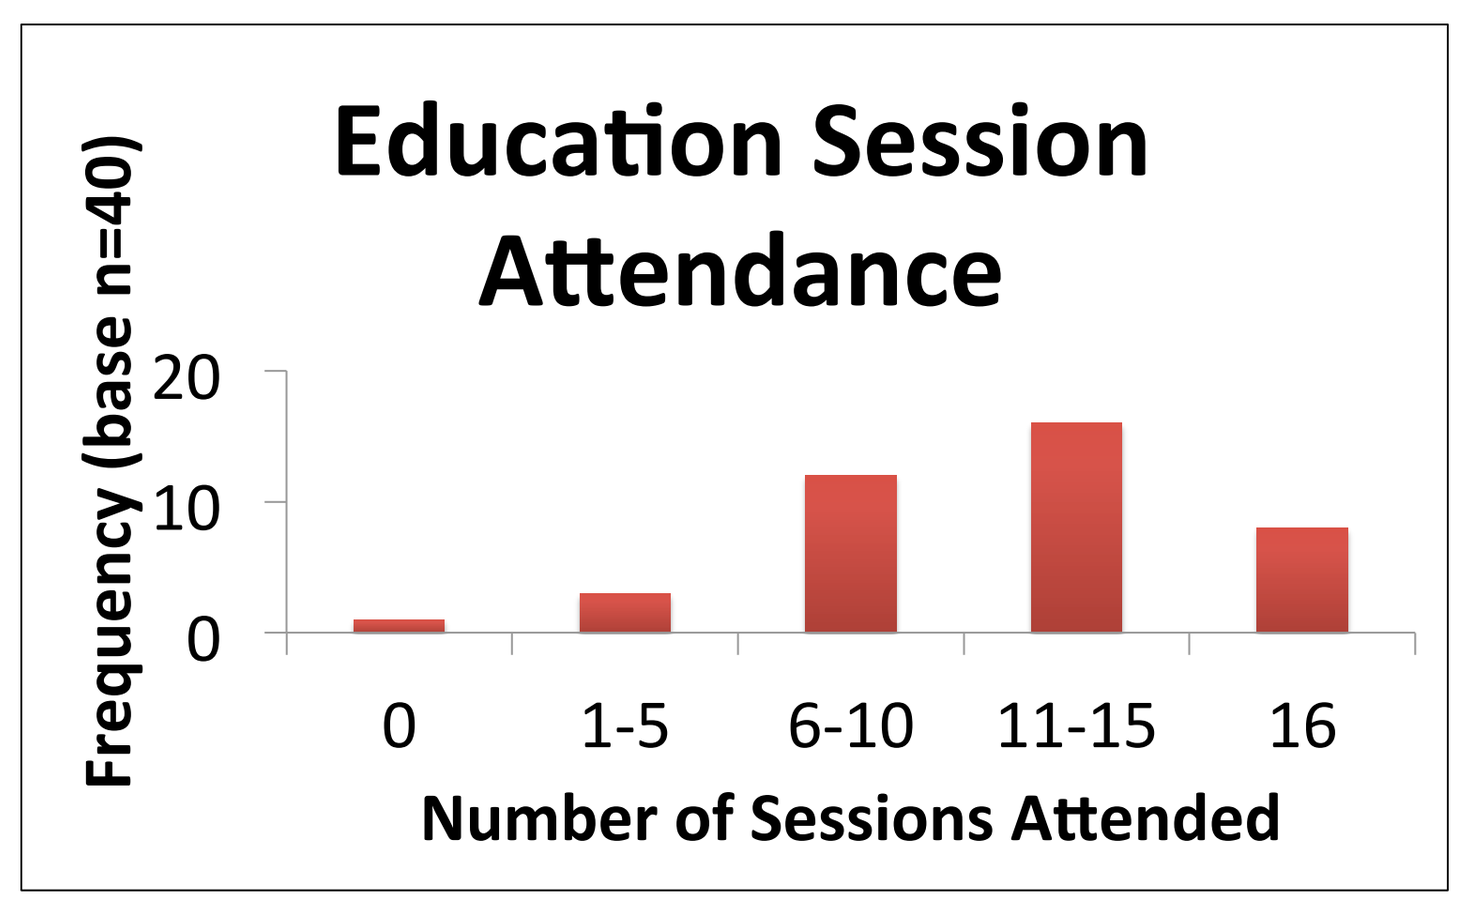
Education Session Attendance
Frequency (base n=40)
0
10
20
0
1-5
6-10
11-15
16
Number of Sessions Attended

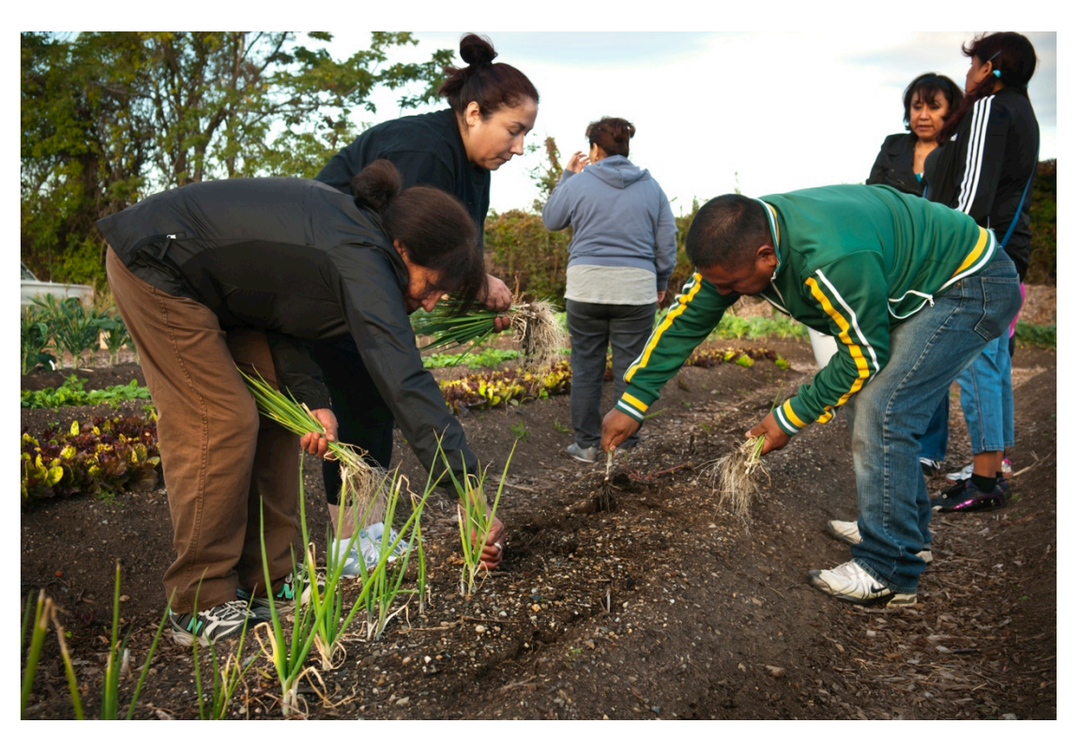
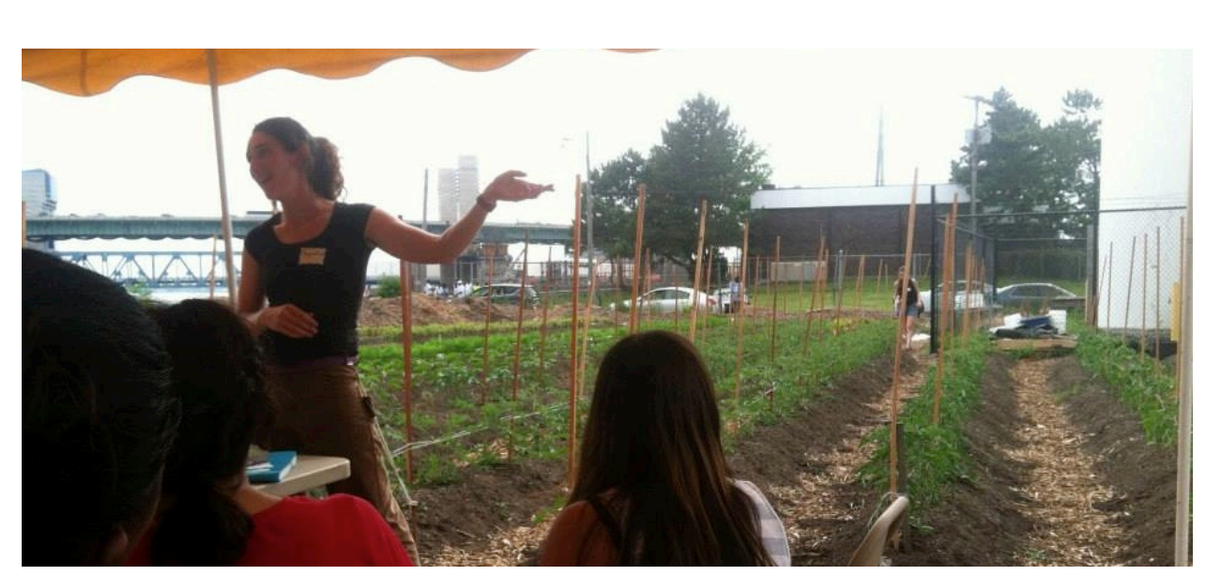

Table 1. Descriptive Statistics for New Haven Farms Membership

Total n=40		Week 0
Age (Mean±SD)		42.1±11.3 y.o.
Race/Ethnicity	Hispanic	77.5%(31)
	African American	10.5%(4)
	Other	12%(5)
Female		90%(36)
Household size (Mean±SD)		3.5±1.8 people
Relationship Status	Married or living with a partner	52.5%(21)
	Single, separated, or divorced	47.5%(19)
At least one child under 5 y.o. in household		35%(14)
Income	Mean±SD	$15,714±$12,256
	Median	$15,000
Education	No High School Degree	40%(16)
	Completed High School	37.5%(15)
	Some College	12.5%(5)
	College Degree	10%(4)
Food Assistance	Total	32.5%(13)
	WIC	7.7%(1)
	SNAP	46.2%(6)
	Food Bank	7.7%(1)
	Unknown	38.5%(5)
Immigration Status	Total Immigrants	70%(28)
	From Mexico	53.6%(15)
	From other Spanish Speaking Countries	46.4%(13)
No. of years in US for immigrant participants (Mean±SD)		18.8±12.7
Spanish Speaking		55%(22)

The sample was primarily female, low-income, Hispanic immigrants, in the US for an average time of 18.8 years. About a third of the group relied on food assistance and had a mean household size of 3.5 people.

Figure 1: Changes in Fruit & Vegetable Consumption

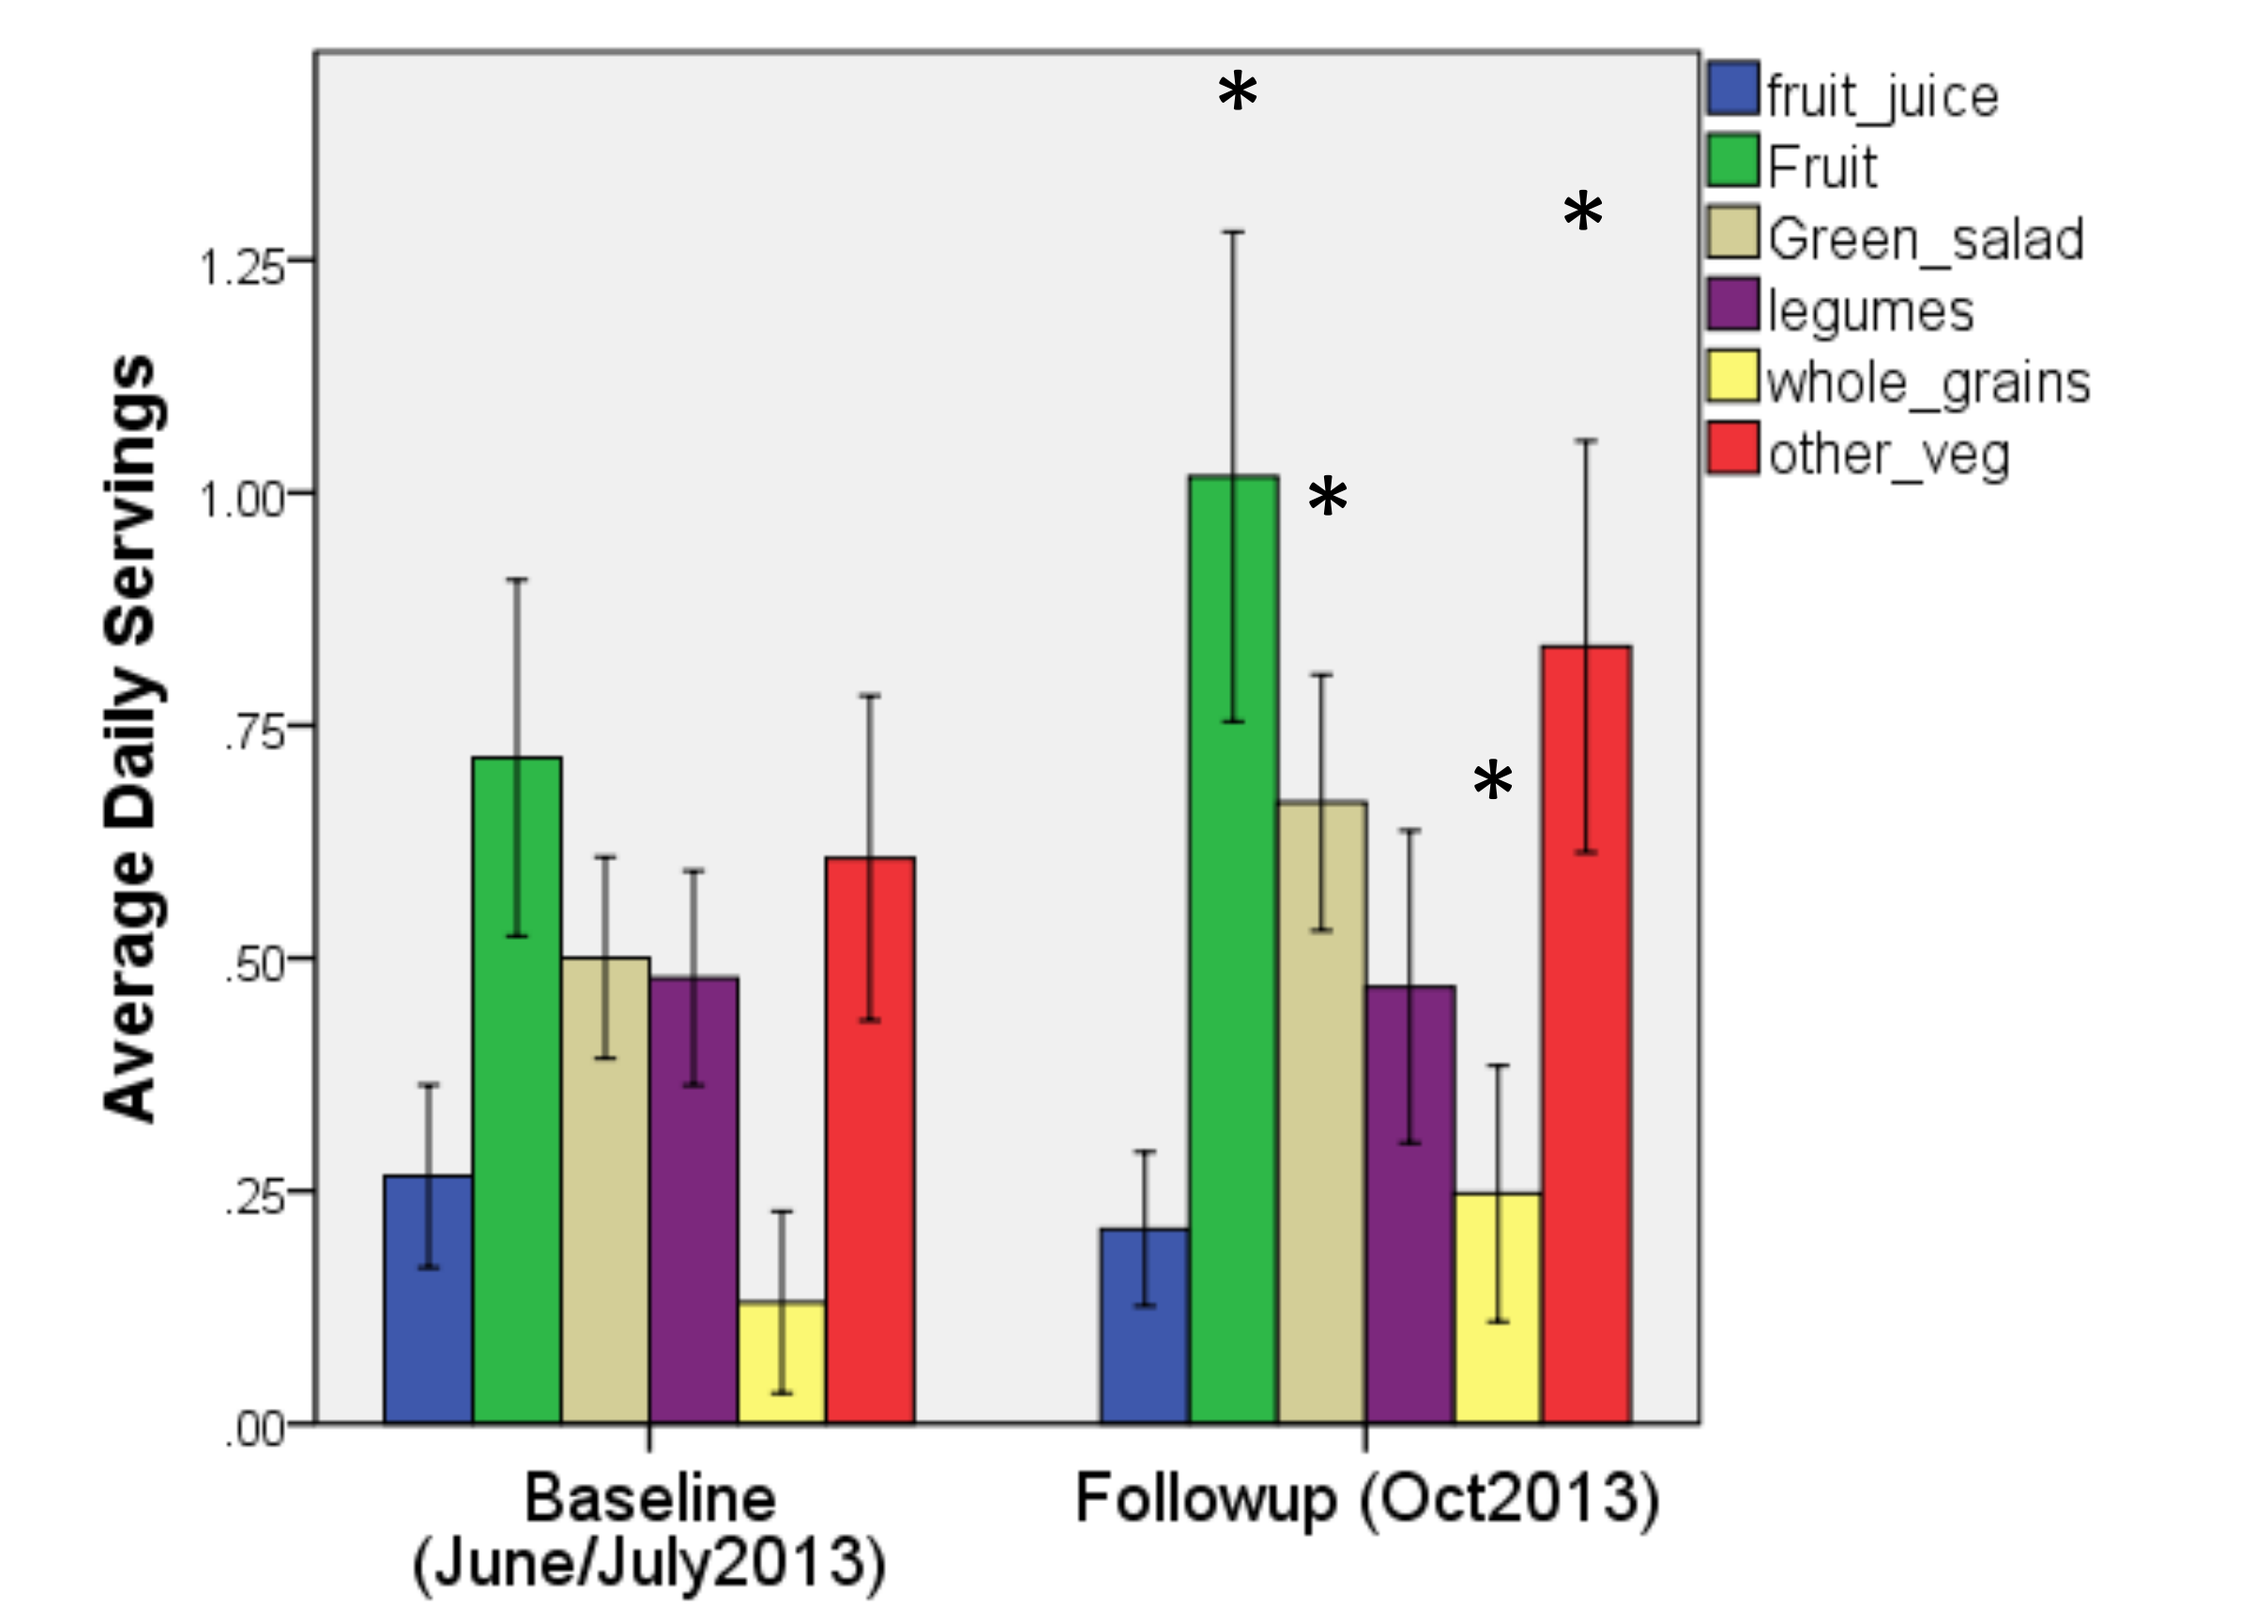
Average Daily Servings
.00
.25
.50
.75
1.00
1.25
Baseline (June/July2013)
Followup (Oct2013)
fruit_juice
Fruit
Green_salad
legumes
whole_grains
other_veg
*

*Significantly Different from baseline at p < 0.05

Error bars: 95% CI

Results: Daily Fruit and Vegetable Consumption (Figure 1)

- Increased daily intake of fruits and vegetables by an average of 0.86 ± 0.25 servings (p=0.002)
- Significantly more daily servings of:
 - Vegetables (+0.263 servings; p=0.036)
 - Whole grains (+0.126 servings; p=.024)
 - Green salad (+0.167 servings; p=0.014)
 - Fruit (+0.31 servings; p=0.034)
- No significant changes in daily servings of any other foods reported (cereal, milk, soda, coffee & tea, fried potatoes, legumes, salsa, pizza, tomatoes, cheese, meat, processed meat, whole grain bread, candy, sweet breads, cake, cookies, frozen desserts or popcorn)

Results: Food Insecurity

According to the Food and Agriculture Organization (FAO), food security "*exists when all people, at all times, have physical and economic access to sufficient, safe and nutritious food to meet their dietary needs and food preferences for an active and healthy life*". (World Food Summit 1996)

Pre and post intervention assessment of food security status shows a:

- 21 percentage point decrease (from 32.4% to 11.4%) with P-value of α = .05 in the proportion of households where adult members needed to cut the size of meals or skip meals because there wasn't enough money for food.
- 22.7 percentage point decrease (from 70.8 % of households to 48.1% of households with P-value of α = .05) in the proportion of member households relying on only a few kinds of low-cost food to feed children because they were running out of money to buy food.
- 26.9 percentage point decrease (from 41.7% to 14.8% with P-value of α = .01) in the proportion of households where children were not eating enough because the family couldn't afford enough food.
- 21.7 percentage point decrease (from 29.1% to 7.4% with P-value of α = .05) in the proportion of households that had to cut the size of the children's meals because there wasn't enough money for food.

Conclusions

Research concerning the dietary and health effects of urban agriculture is scarce. This pilot program aimed to test the impact of local access to high quality fruits and vegetables (F&V) in combination with a nutrition and cooking education component that supports F&V intake.

- New Haven Farms Fresh Produce Prescription Program increased fruit and vegetable consumption by .86 servings per day for individual members.
- F&V consumption of other members of the household may also have increased, as food tends to be shared within the household.
- Food insecurity was reduced in both adults who took part in the program as well as in the children who reside with them.

Further research is necessary to better understand the mechanisms, duration of impact, cost-effectiveness, as well as dose-response effect. The size of the study and the ability to test the intervention on other populations will inform how such programs could be scaled up to have broader population effects aimed at nutrition related diabetes risk.

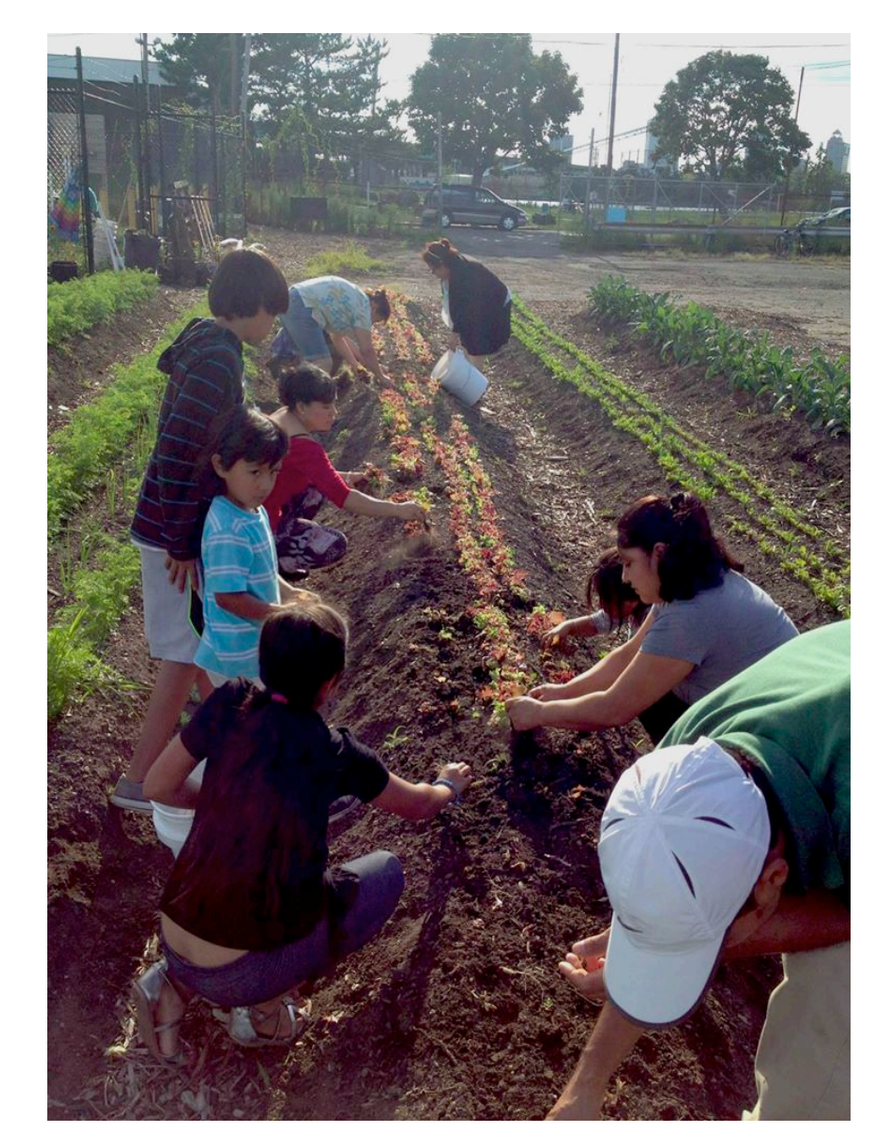

References

Anderson JV, Bybee DI, Brown RM, McLean DF, Garcia EM, Breer L, Schillo BA. (2001) 5 A Day fruit and vegetable intervention improves consumption in a low-income population. J Am Diet Assoc 101(2): 195-202.
Katherine Alaimo, PhDemail address, Elizabeth Packnett, MPH, Richard A. Miles, BS, Daniel J. Kruger. "Fruit and Vegetable Intake among Urban Community Gardeners" Journal of Nutrition Education and Behavior. Vol0, 2 , Pages 94-101, March 2008
Cohen, N.L., J.P. Cooley, R.B. Hall and AmM. Stoddard (1997). "Community suported agriculture: A study of share-holders' dietary patterns and food practices." Paper read at the International conference on Agricultural Production and Nutrition . March, At Tufts University, School of Nutrition Science and Policy, Boston, MA)
Jill S. Litt, Mah-J. Soobader, Mark S. Turbin, James W. Hale, Michael Buchenau, Julie A. Marshall. American Journal of Public Health: August 2011, Vol. 101, No. 8: 1466–1473.
Johnson DB, Beaudoin S, Smith LT, Beresford SAA, LoGerfo JP. Increasing fruit and vegetable intake in homebound elders: the Seattle Senior Farmers' Market Nutrition Pilot Program. Prev Chronic Dis [serial online] 2004 Jan [date cited]. Available from: URL: http://www.cdc.gov/pcd/issues/2004/jan/03_00010a.htm
Sarah Wakefield, Fiona Yeudall, Carolin Taron, Jennifer Reynolds and Ana Skinner. "Growing urban health: Community gardening in South-East Toronto". Health Promotion International, Vol. 22 No. 2
World Food Summit 1996, Rome Declaration on World Food Security.

Acknowledgements

The authors thank New Haven Farms' Board of Directors, volunteers and staff: W. Albandoz, J. Appel, L. Audette, S. Aybar, J. Bishop, M. Burton, A. Camp, E. Caldwell, P. Dillon, C. Duckwall, A. Chojolan, N. Dennett, R. Freidman, A. Florecita, C. Granda, J. Greenberg, P. Horta, A. Johnson, N. Kline, B. Lai, J. Maisonpierre, E. Magenheimer, E. Marshall, D. Medina, C. Metcalf, M. Centrone-Stefani, A. Montosa, C. Negaro, M. Newman, A. Paine, P. Perez, M. Perez, J. Rappaport, J. Reyes, J. Romero, M. Smith, N. Smith, E. Swenson, G. Veitch, M. Walsh. Thank you to the CT Dept of Agriculture for funding support.